Prolonged Plasma Urate-Lowering after a Single Intravenous Administration of PRX-115, a Novel PEGylated Uricase, in Participants with Elevated Urate Levels

Christian Schwabe[1], **Orit Cohen-Barak**[2], Alexandra Cole[3], Hadar Reuveni[2], Liron Shelev[2], Liora Blinder-Haddad[2] and Nicola Dalbeth[4].

[1]Schwabe, Christian, MD, New Zealand Clinical Research, Auckland, NZ [2]Orit Cohen-Barak, PhD, Hadar Reuveni, Liron Shelev and Liora Blinder-Haddad, Protalix Ltd., Carmiel, Israel [3]Alexandra Cole MBChB, FRNZCGP, DHP, New Zealand Clinical Research, Christchurch, NZ
[4]Nicola Dalbeth, MBChB, MD, FRACP, FRSNZ, University of Auckland, NZ

BACKGROUND

Gout is a common and painful inflammatory condition caused by elevated urate levels in the blood, leading to the deposition of urate crystals in joints and tissues. Uricase catalyzes the breakdown of urate into allantoin, a more soluble and easily excreted compound, thereby reducing urate levels in the blood. PRX-115 is a recombinant homotetrameric uricase produced from *Candida utilis* using proprietary plant-based protein expression system. The enzyme is uniquely modified post purification with specific polyethylene glycol (PEG) molecules. PRX-115 is designed to improve stability and increase half-life, reduce immunogenicity, and maintain high specific activity. The unique chemical modifications aim to enhance efficacy and safety, making PRX-115 a potential new treatment for gout.

METHODS

A first-in-human (FIH) single ascending dose (SAD), double-blinded, placebo-controlled study (FIH-SAD, NCT05745727) was conducted to assess the safety, tolerability, pharmacokinetics (PK), and pharmacodynamics (PD) of PRX-115 following a single intravenous (IV) infusion in subjects with elevated plasma urate levels. The study recruited 64 subjects in 8 cohorts, largely with elevated urate (>6.0 mg/dL), randomized to PRX-115 or placebo in a 3:1 ratio (6 subjects on active and 2 on placebo per cohort). Subjects were monitored for 85 days (12 weeks) post-infusion. See **Figure 1** for a graphic presentation of the study scheme and study schedule per subject. Validated methods were developed for assessing PRX-115 plasma concentrations for PK evaluation, presence of serum anti-drug antibody (ADA), and plasma urate levels. PRX-115 concentrations and plasma urate levels were measured frequently throughout the study, up to 12 weeks following the single IV infusion.

Figure 1: Study Scheme and Study Schedule Per Subject

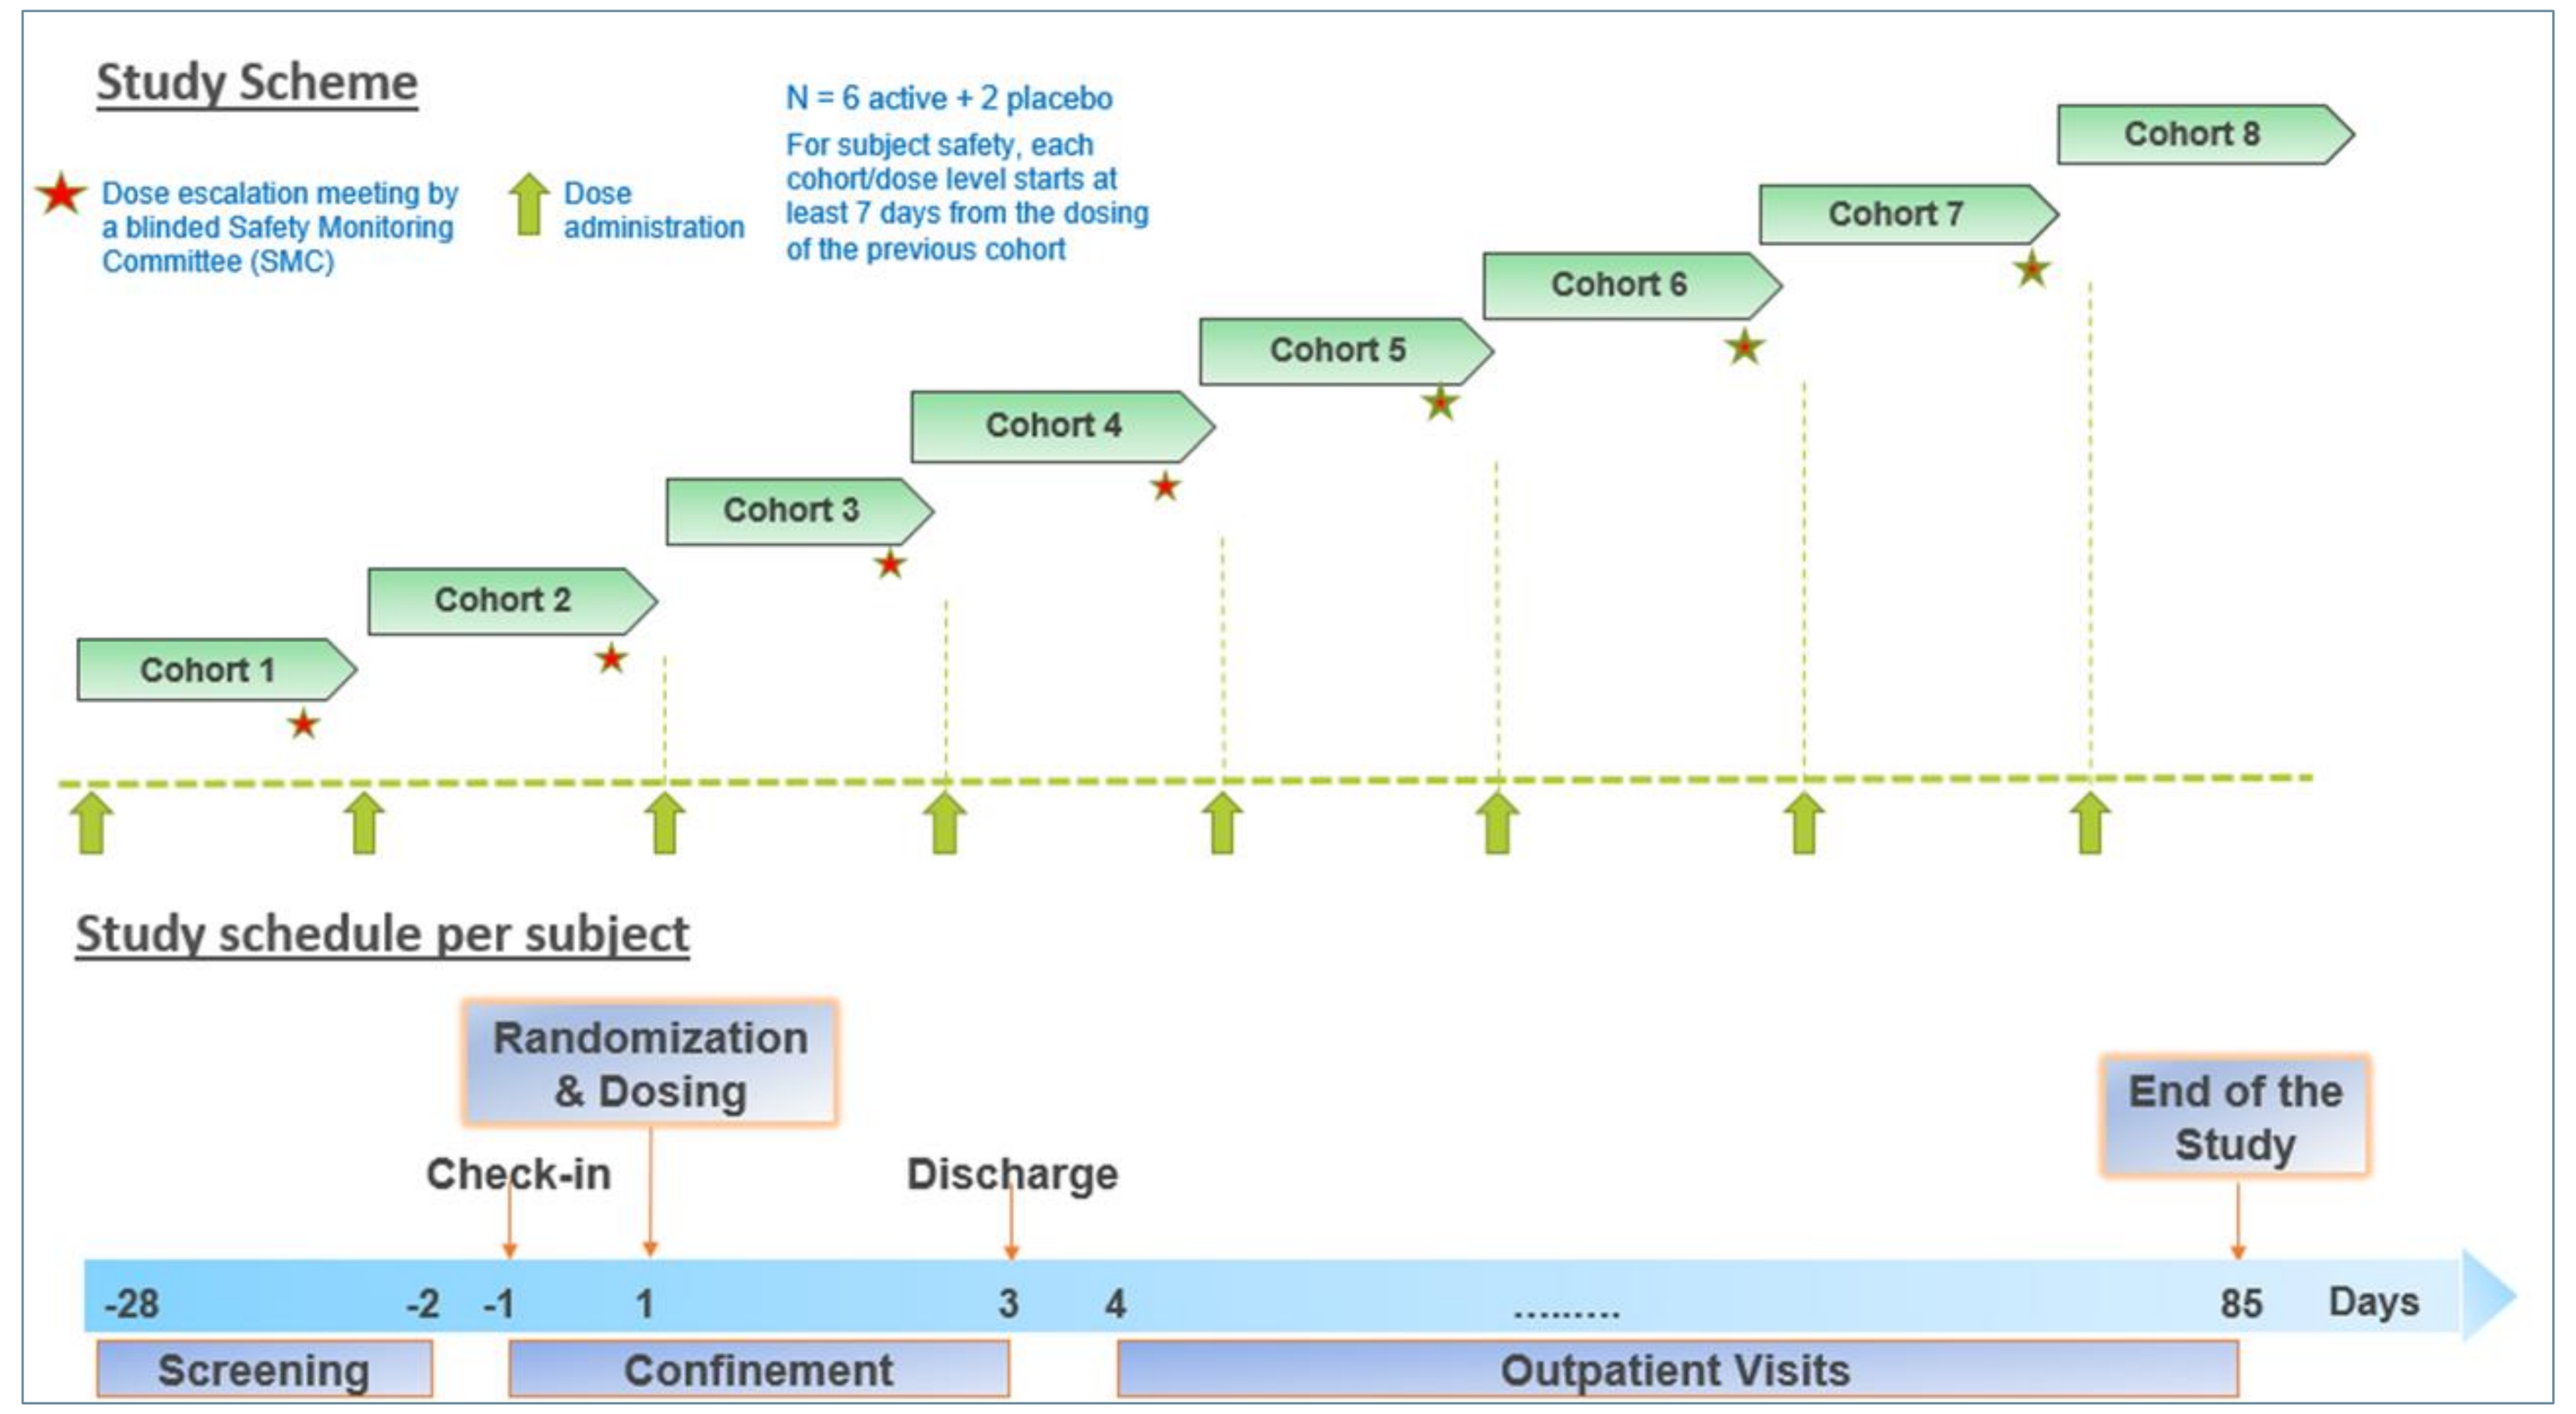
Study Scheme
Dose escalation meeting by a blinded Safety Monitoring Committee (SMC)
Dose administration
N = 6 active + 2 placebo
For subject safety, each cohort/dose level starts at least 7 days from the dosing of the previous cohort
Cohort 1
Cohort 2
Cohort 3
Cohort 4
Cohort 5
Cohort 6
Cohort 7
Cohort 8
Study schedule per subject
Randomization & Dosing
Check-in
Discharge
End of the Study
-28
-2
-1
1
3
4
.........
85
Days
Screening
Confinement
Outpatient Visits

RESULTS

Demographics

The FIH SAD study included 64 Subjects: 48 received PRX-115 in different dose cohorts and 16 received placebo and were pooled for the analysis. See **Table 1** for Demographics and Baseline Characteristics.

Table 1: Demographics and Baseline Characteristics

Parameter	Statistic	Pooled PRX-115	Pooled Placebo	Overall
	n	48	16	64
Age (years)	Mean	35.9	33.4	35.3
	SD	12.3	10.8	11.9
Sex n(%)	Female	16(33.3)	5(31.3)	21(32.8)
	Male	32(66.7)	11(68.8)	43(67.2)
Race n(%)	American Indian or Alaska Native	0	0	0
	Asian	7(14.6)	5(31.3)	12(18.8)
	Black or African American	1(2.1)	0	1(1.6)
	Native Hawaiian or other Pacific Islander	8(16.7)	2(12.5)	10(15.6)
	White	31(64.6)	8(50.0)	39(60.9)
	Other	3(6.3)	2(12.5)	5(7.8)
Weight (kg)	Mean	87.52	85.87	87.11
	SD	18.06	18.37	18.01
Body Mass Index (kg/m^2)	Mean	29.16	28.01	28.87
	SD	5.03	5.35	5.10
Plasma Urate (mg/dL)	Mean per cohort	7.0-8.5	7.0	7.0-8.5

Safety and Tolerability

PRX-115 was found to be well tolerated. All randomized subjects completed the study. The total number of Treatment Emergent Adverse Event (TEAE) were similar in the active treatment groups and placebo (~80%). Only 25% of the subjects treated with PRX-115 (12/48) reported study drug related AEs. The majority of these AEs were mild to moderate and transient in nature. One subject, in cohort 2, experienced an anaphylactic reaction immediately following the start of the infusion (6 min), and thus was exposed to 5% of the applicable PRX-115 dose. The reaction resolved completely and the subject remained in the study for follow-up safety assessments. Following this anaphylaxis event, premedication of anti-histamines and steroids were administered to all subjects from that point forward. No other subjects experienced a similar reaction, and no other serious adverse events (SAEs) were reported during the study (See **Table 2** for summary of AEs).

Table 2: Overall Summary of Adverse Events*

	Cohort 1	Cohort 2	Cohort 3	Cohort 4	Cohort 5	Cohort 6	Cohort 7	Cohort 8	Pooled PRX-115	Pooled Placebo
N	6	6	6	6	6	6	6	6	48	16
TEAE n(%)	5(83.3)	6(100.0)	5(83.3)	3(50.0)	6(100.0)	5(83.3)	3(50.0)	4(66.7)	37(77.1)	13(81.3)
Related TEAE n(%)	1(16.7)	5(83.3)	3 (50.0)	1(16.7)	1(16.7)	0	0	1(16.7)	12(25.0)	3(18.8)
Serious Related TEAE n(%)	0	1(16.7)	0	0	0	0	0	0	1(2.4)	0
TEAE Leading to Study Drug Discontinuation n(%)	0	1(16.7)	0	0	0	0	0	0	1(2.1)	0
TEAE Leading to Study Discontinuation n(%)	0	0	0	0	0	0	0	0	0	0

*Number of Subjects reporting at least one adverse event
TEAE= Treatment Emergent Adverse Event

Pharmacokinetics

PRX-115 plasma concentrations are graphically presented in **Figure 2** for all cohorts. PRX-115 exposure increased in a dose–dependent manner with maximal concentrations observed, in general, immediately post-infusion, and detectable PRX-115 levels were observed in plasma for up to 12 weeks from subjects in cohorts 6, 7 and 8.

Figure 2: PRX-115 Plasma Concentrations Over Time

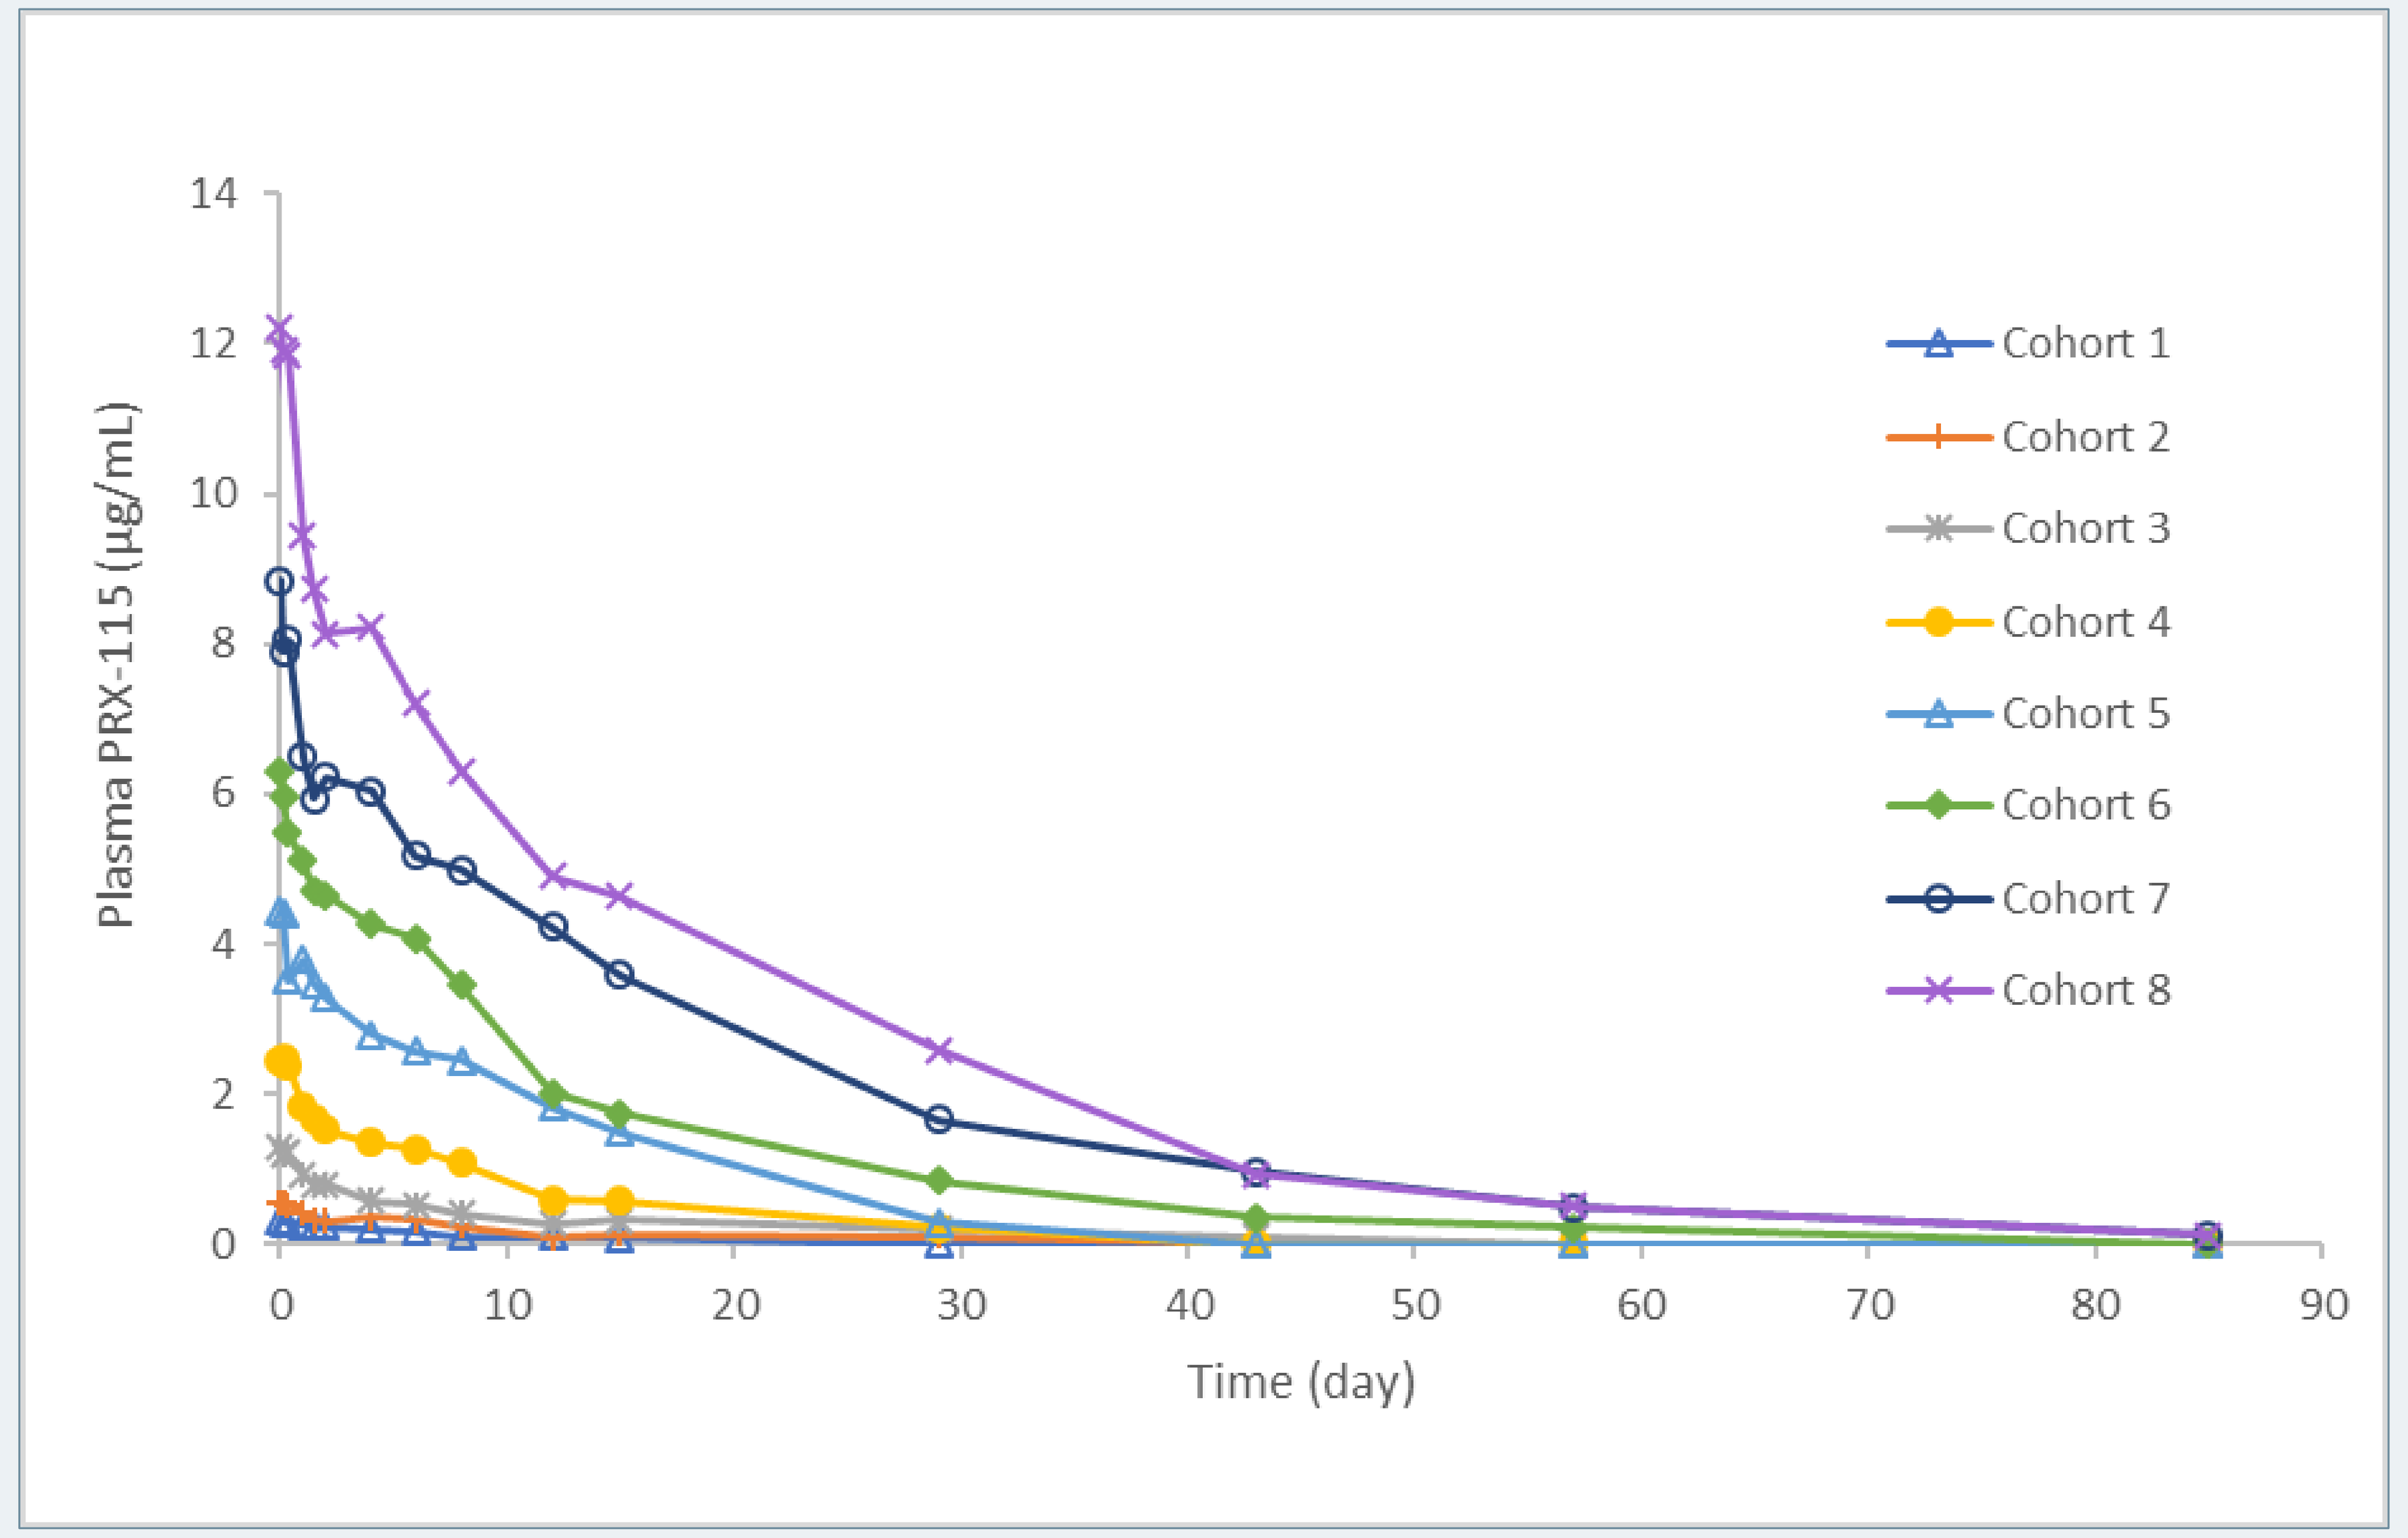
Plasma PRX-115 (µg/mL)
Time (day)
Cohort 1
Cohort 2
Cohort 3
Cohort 4
Cohort 5
Cohort 6
Cohort 7
Cohort 8

Pharmacodynamics

In all tested doses, a single dose of PRX-115 rapidly reduced plasma urate levels. The effect and duration of response were dose-dependent. Following a single dose, mean plasma urate levels remained below 6.0 mg/dL for up to 12 weeks at the highest dose levels (**Figure 3**).

Figure 3: Plasma Urate Concentrations Over Time

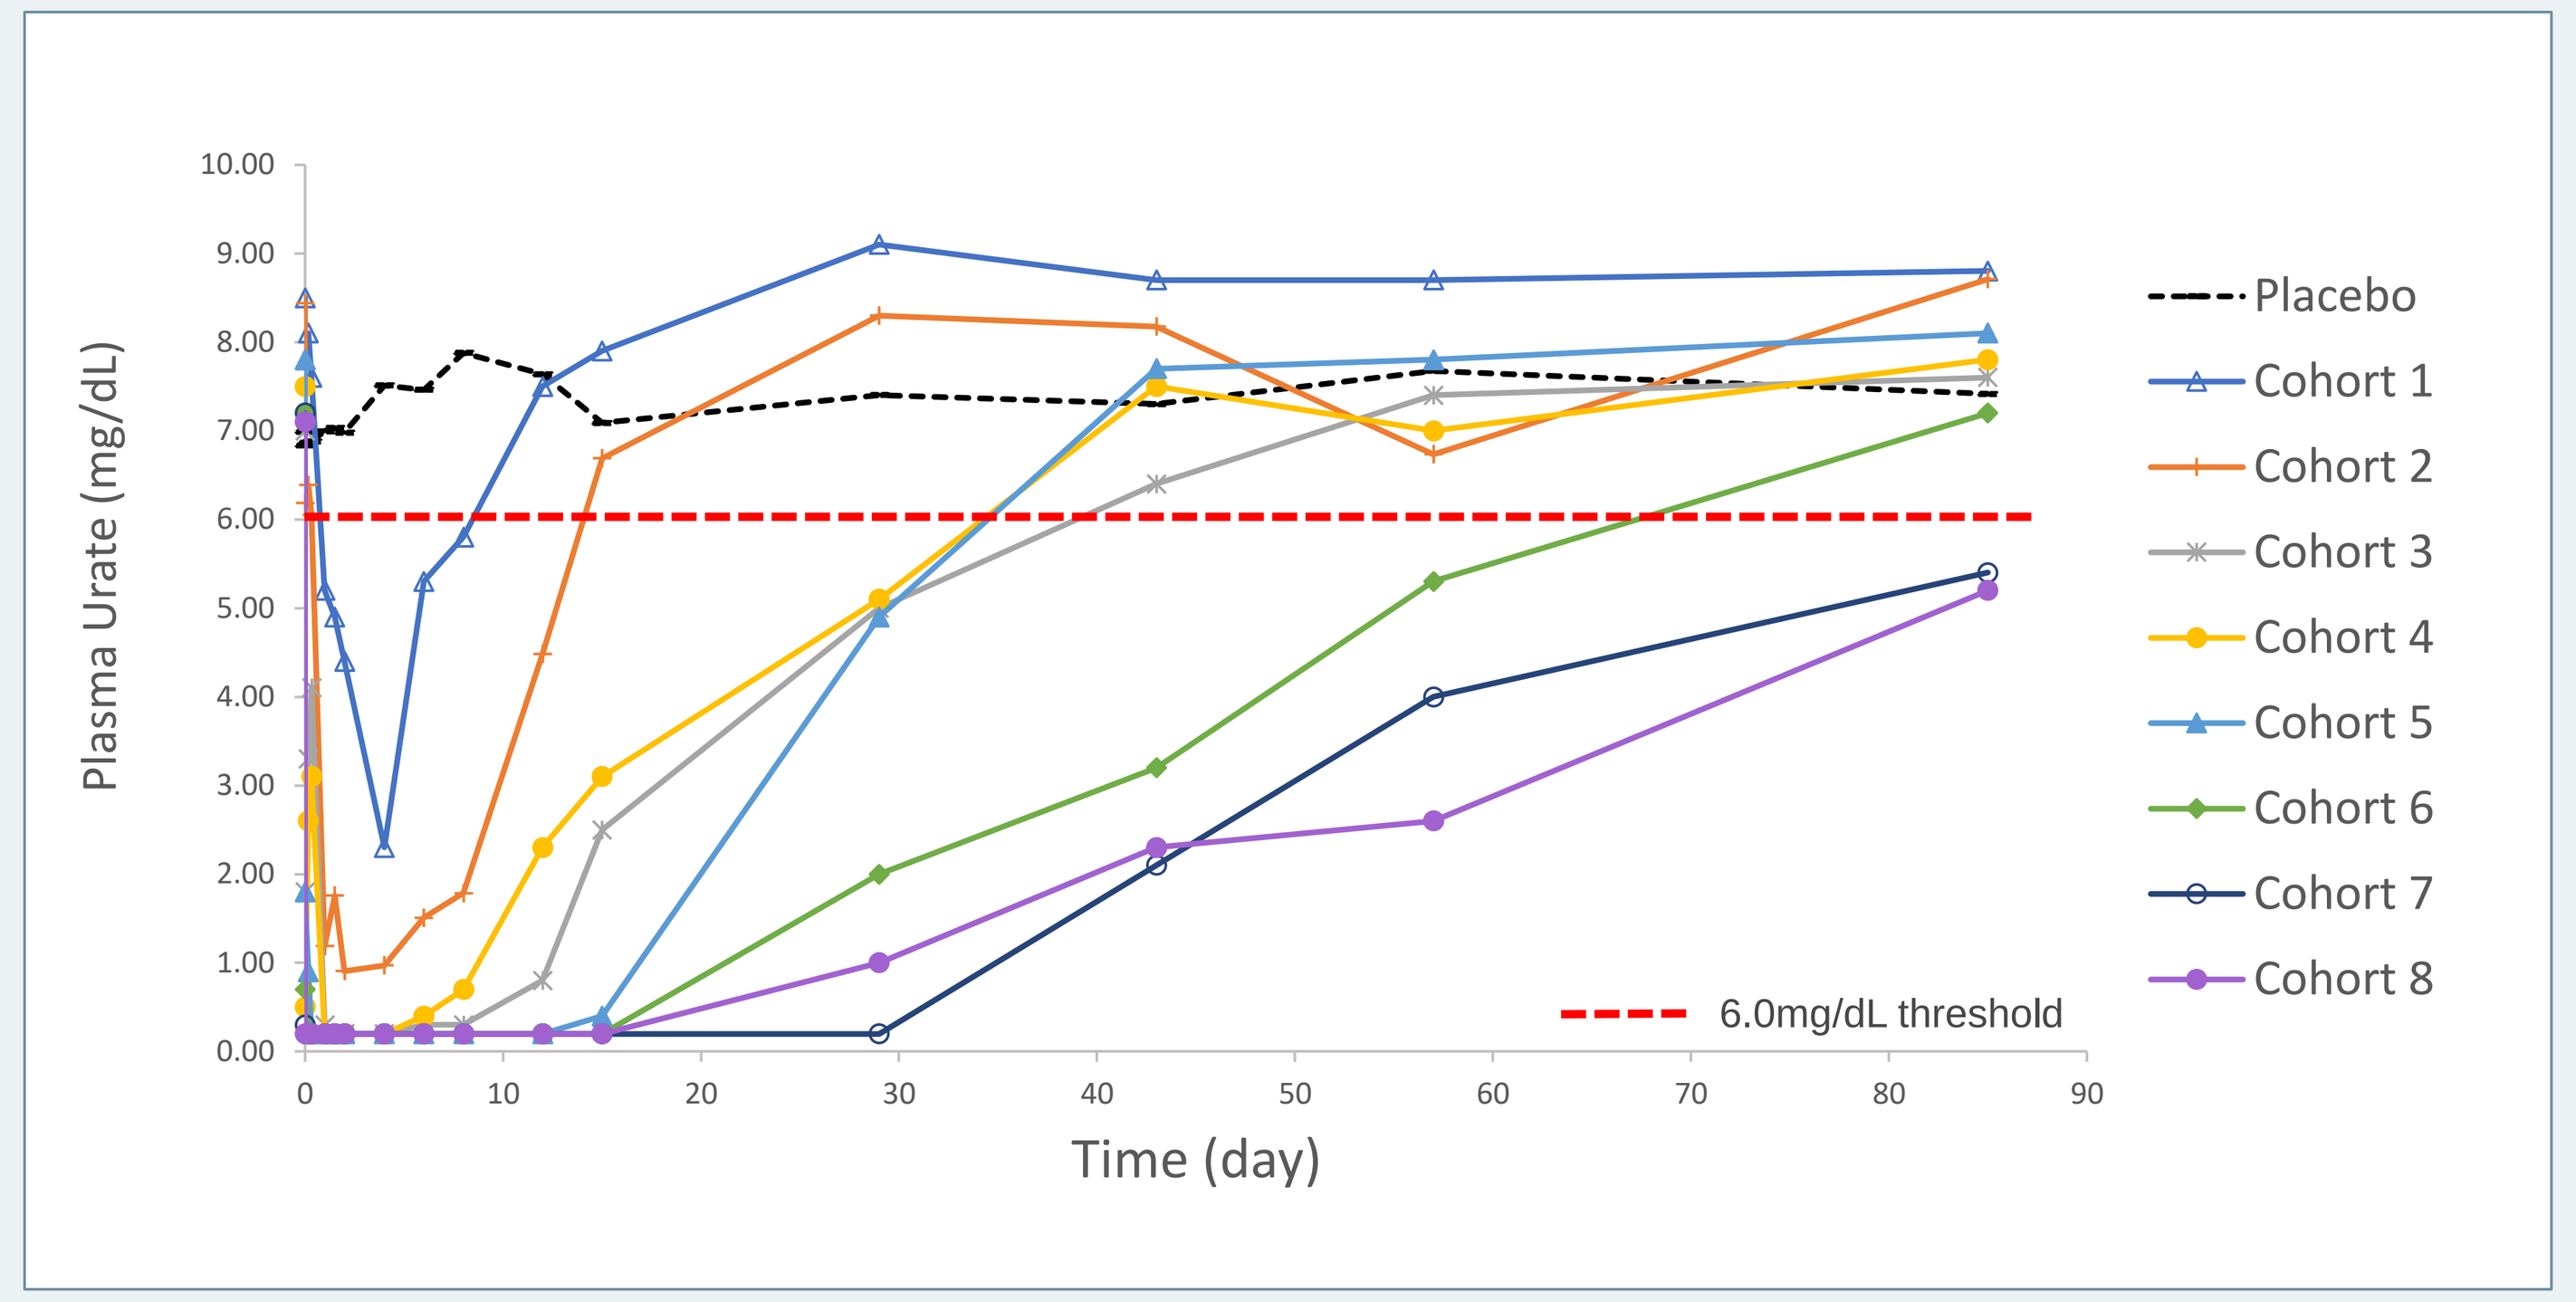
Plasma Urate (mg/dL)
Time (day)
Placebo
Cohort 1
Cohort 2
Cohort 3
Cohort 4
Cohort 5
Cohort 6
Cohort 7
Cohort 8
6.0mg/dL threshold

CONCLUSIONS

The findings from this First-in-Human (FIH) Single Ascending Dose (SAD) study, involving 8 ascending dose cohorts, propose compelling evidence of prolonged PRX-115 exposure with corresponding prolonged duration of effect on plasma urate levels.

PRX-115 was safe and well tolerated following single IV doses.

The study results suggest that PRX-115 may offer an effective urate-lowering treatment with an added benefit of a potentially wide dosing interval, which may enhance patient compliance and treatment flexibility.

DISCLOSURES

C. Schwabe: None; **O. Cohen Barak**: Protalix Ltd, 3,; **A. Cole**: None; **H. Reuveni**: Protalix Ltd, 3; **L. Shelev**: Protalix Ltd, 3; **L. Blinder-Haddad**: Protalix Ltd, 3, 11; **N. Dalbeth**: Protalix, 2.